Creating the Next Generation of Nutrition Leaders in Uganda: The Uganda Nutrition Fellowship (UNF) Program

Bridget Ralph, Brenda Namugumya, Dr. Hanifa Bachou, Karen Tumwine, Rebecca Namara, and Kristen Cashin, FHI 360/Food and Nutrition Technical Assistance III Project (FANTA)

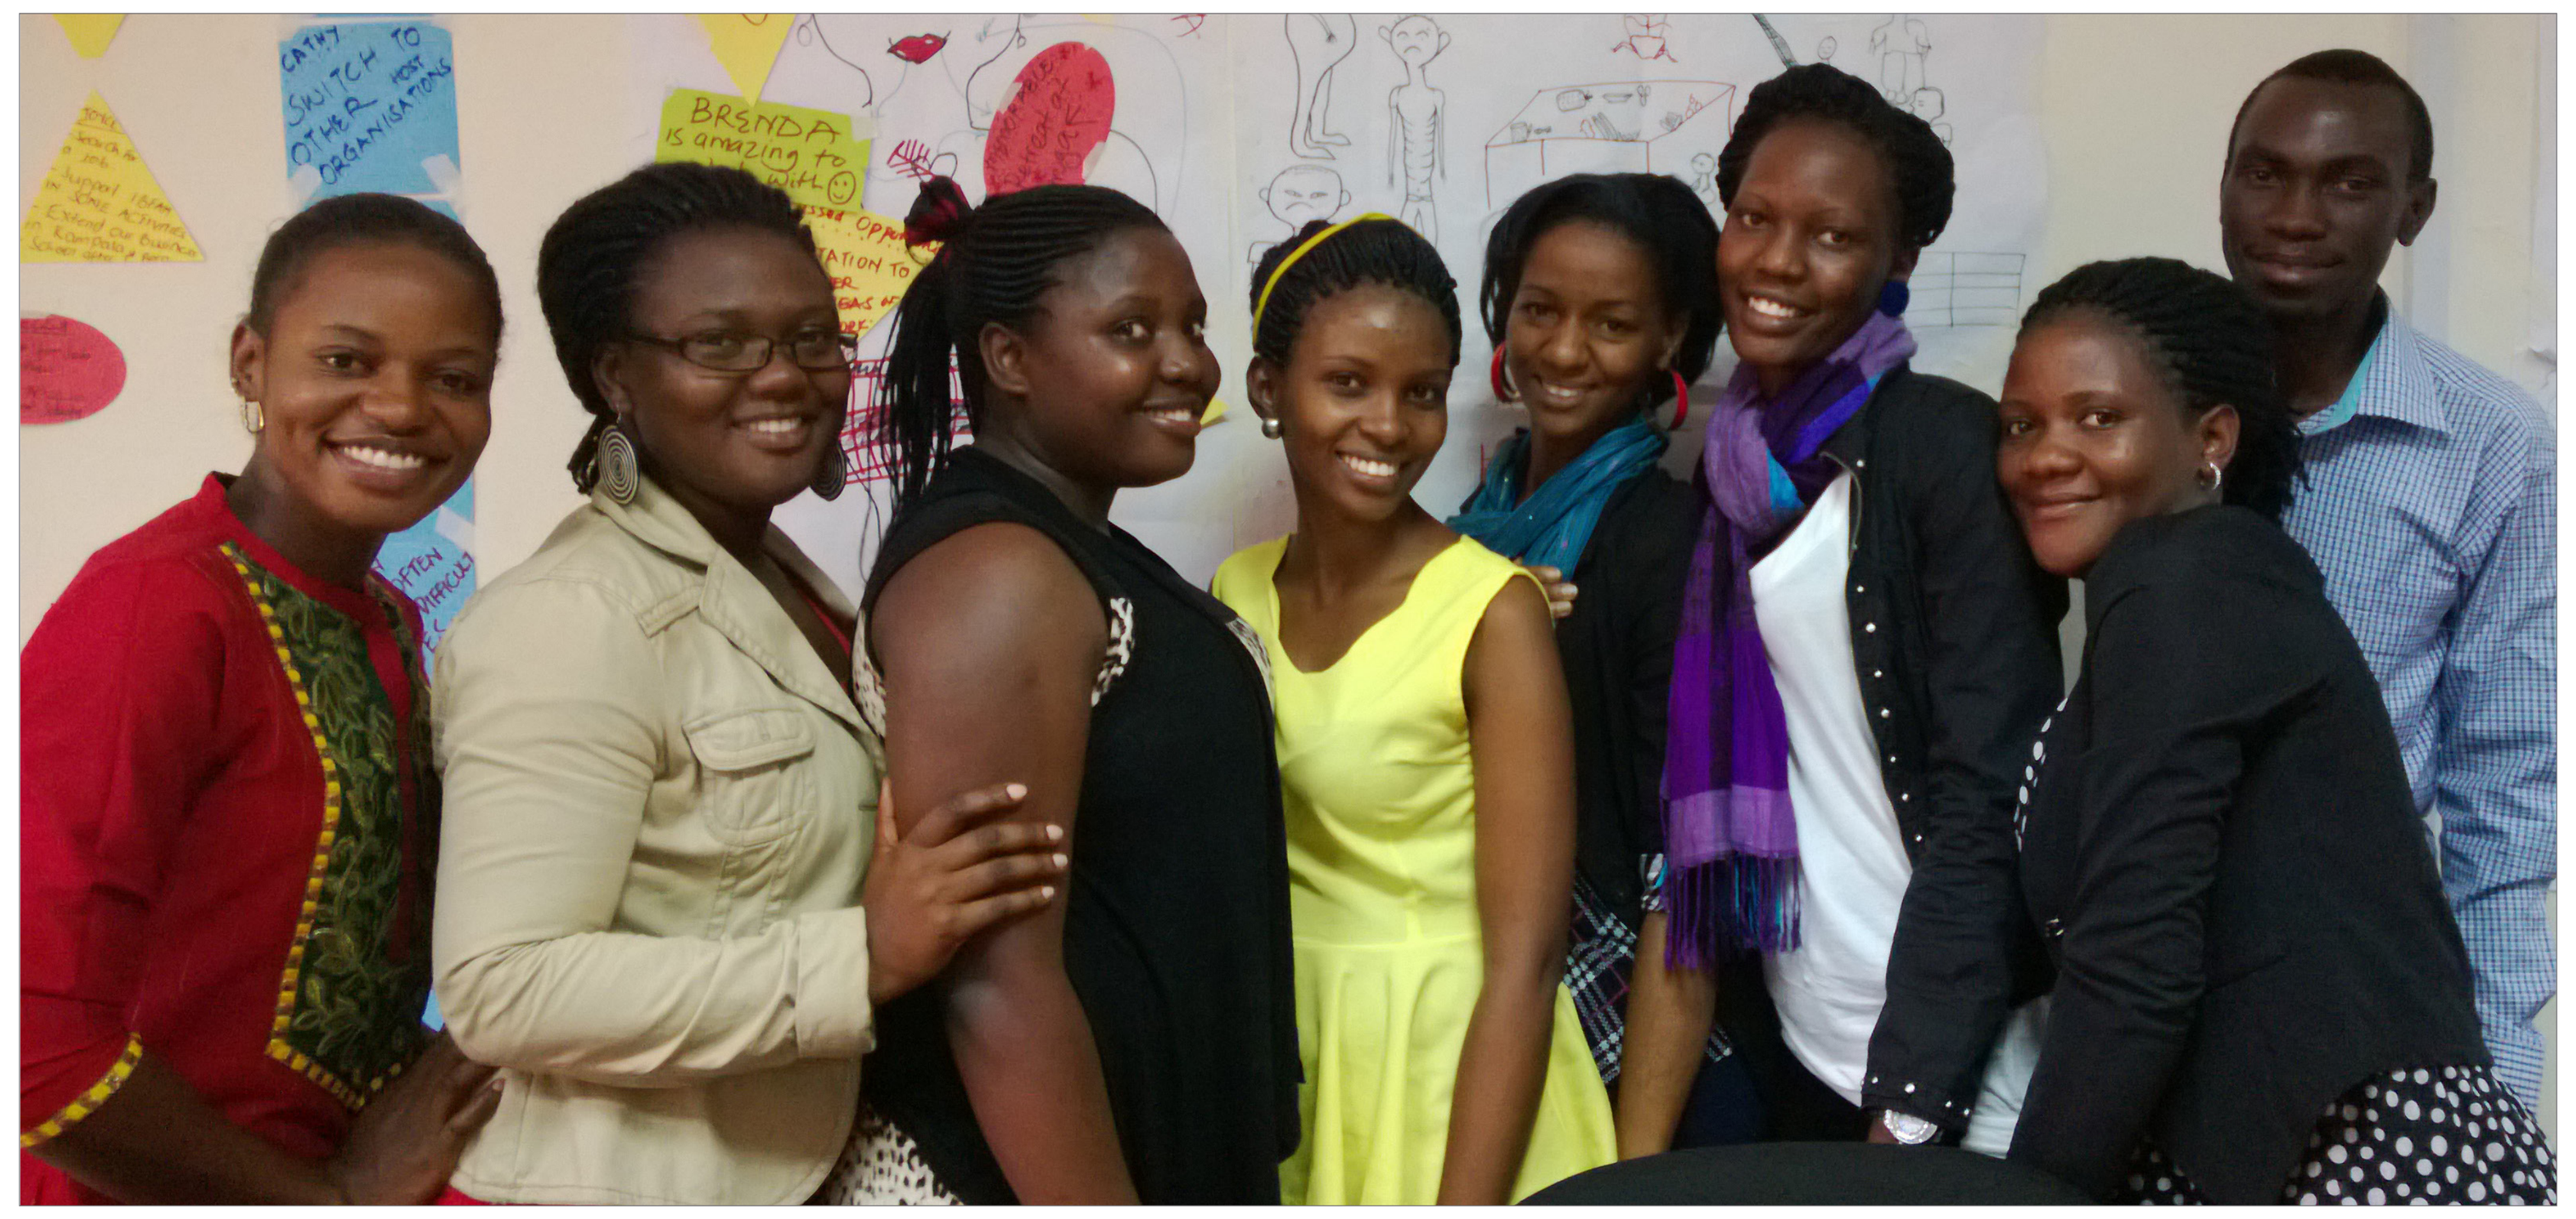

UNF fellows 2013–2014

INTRODUCTION

Addressing Uganda's "double burden" of malnutrition (underweight and overweight) requires strong nutrition leadership and technical skill from the national to community level. Universities provide Uganda's graduates with a theoretical foundation in nutrition, but little practical experience or training on implementation skills such as leadership, communication, work planning, project management, and monitoring, all of which are essential for establishing nutrition goals and objectives and achieving them on time and on budget. To strengthen these skills, the Food and Nutrition Technical Assistance III Project (FANTA), funded by the U.S. Agency for International Development (USAID), in collaboration with stakeholders, initiated the Uganda Nutrition Fellowship (UNF) program in June 2013 for graduates with degrees in nutrition.

PROGRAM OBJECTIVES

- Develop skilled nutrition practitioners by providing a unique mix of practical experience, professional development, leadership training, experience sharing, technical training, and mentorship.
- Provide host organizations with highly motivated and well-qualified young professionals to support their nutrition work.

METHODS

Fellows were placed in government, nongovernmental organizations, and clinical facilities for 12 months. They received support and mentorship from FANTA's UNF staff as well as their host organization mentor and supervisor. Fellows received monthly stipends based on the salary of an entry level government nutritionist and were required to submit monthly reports and timesheets. At their host organizations fellows engaged in policy development, nutrition governance, case management, capacity building, social and behavior change, and community mobilization with guidance from mentors and supervisors at their host organizations. The UNF hosted workshops and retreats for fellows which provided professional development in communication, management, leadership, and team building—skills needed for the next generation of leaders.

Using their newly-refined skills, fellows completed an independent project that addressed a programmatic, policy, or practical challenge observed during their fellowship. UNF projects included topics such as promoting breastfeeding in emergency situations, standardizing health and nutrition messages for health care workers at hospitals, and operationalizing district level governance to improve nutrition. These projects gave fellows an opportunity to take a critical look at their work environment, identify and research an area that could be improved upon, and make recommendations to their host organization.

YEAR ONE PROGRAM DETAILS

In the first year of the UNF program 8 fellows were selected out of 65 applicants. These fellows were selected based on school performance, personal statements, and a panel interview. Of 17 organizations that submitted applications to host a fellow, 8 were selected based on willingness to provide supervision, mentorship, and an educational and meaningful experience. Host organizations included the Uganda Ministry of Health (Nutrition Unit and AIDS Control Program), two USAID projects: Community Connector and Strengthening Partnerships Results and Innovations in Nutrition Globally, the International Baby Food Action Network in Uganda, World Vision Uganda, Nyakibale Hospital, and the Mwanamugimu Nutrition Unit at Mulago Hospital.

The UNF hosted three retreats and two professional development workshops throughout the year. The retreats were an opportunity for the fellows to hear technical presentations on various topics from experts in the field, to work on their UNF projects and posters, and to discuss experiences and share best practices among each other and with UNF staff. The professional development workshops were designed to be informative and interactive. The first workshop focused on communications and leadership and provided an opportunity for fellows to apply what they had learned in working group sessions. The second workshop focused on writing and provided a forum for the fellows to learn about professional writing, research, literature reviews, and citing sources in the context of their UNF project reports.

Host organization mentors and supervisors played a key role in the fellowship experience. They provided guidance and support as fellows navigated the workforce for the first time and served as resources to support fellows on work assignments, identifying UNF project topics, and answering career questions.

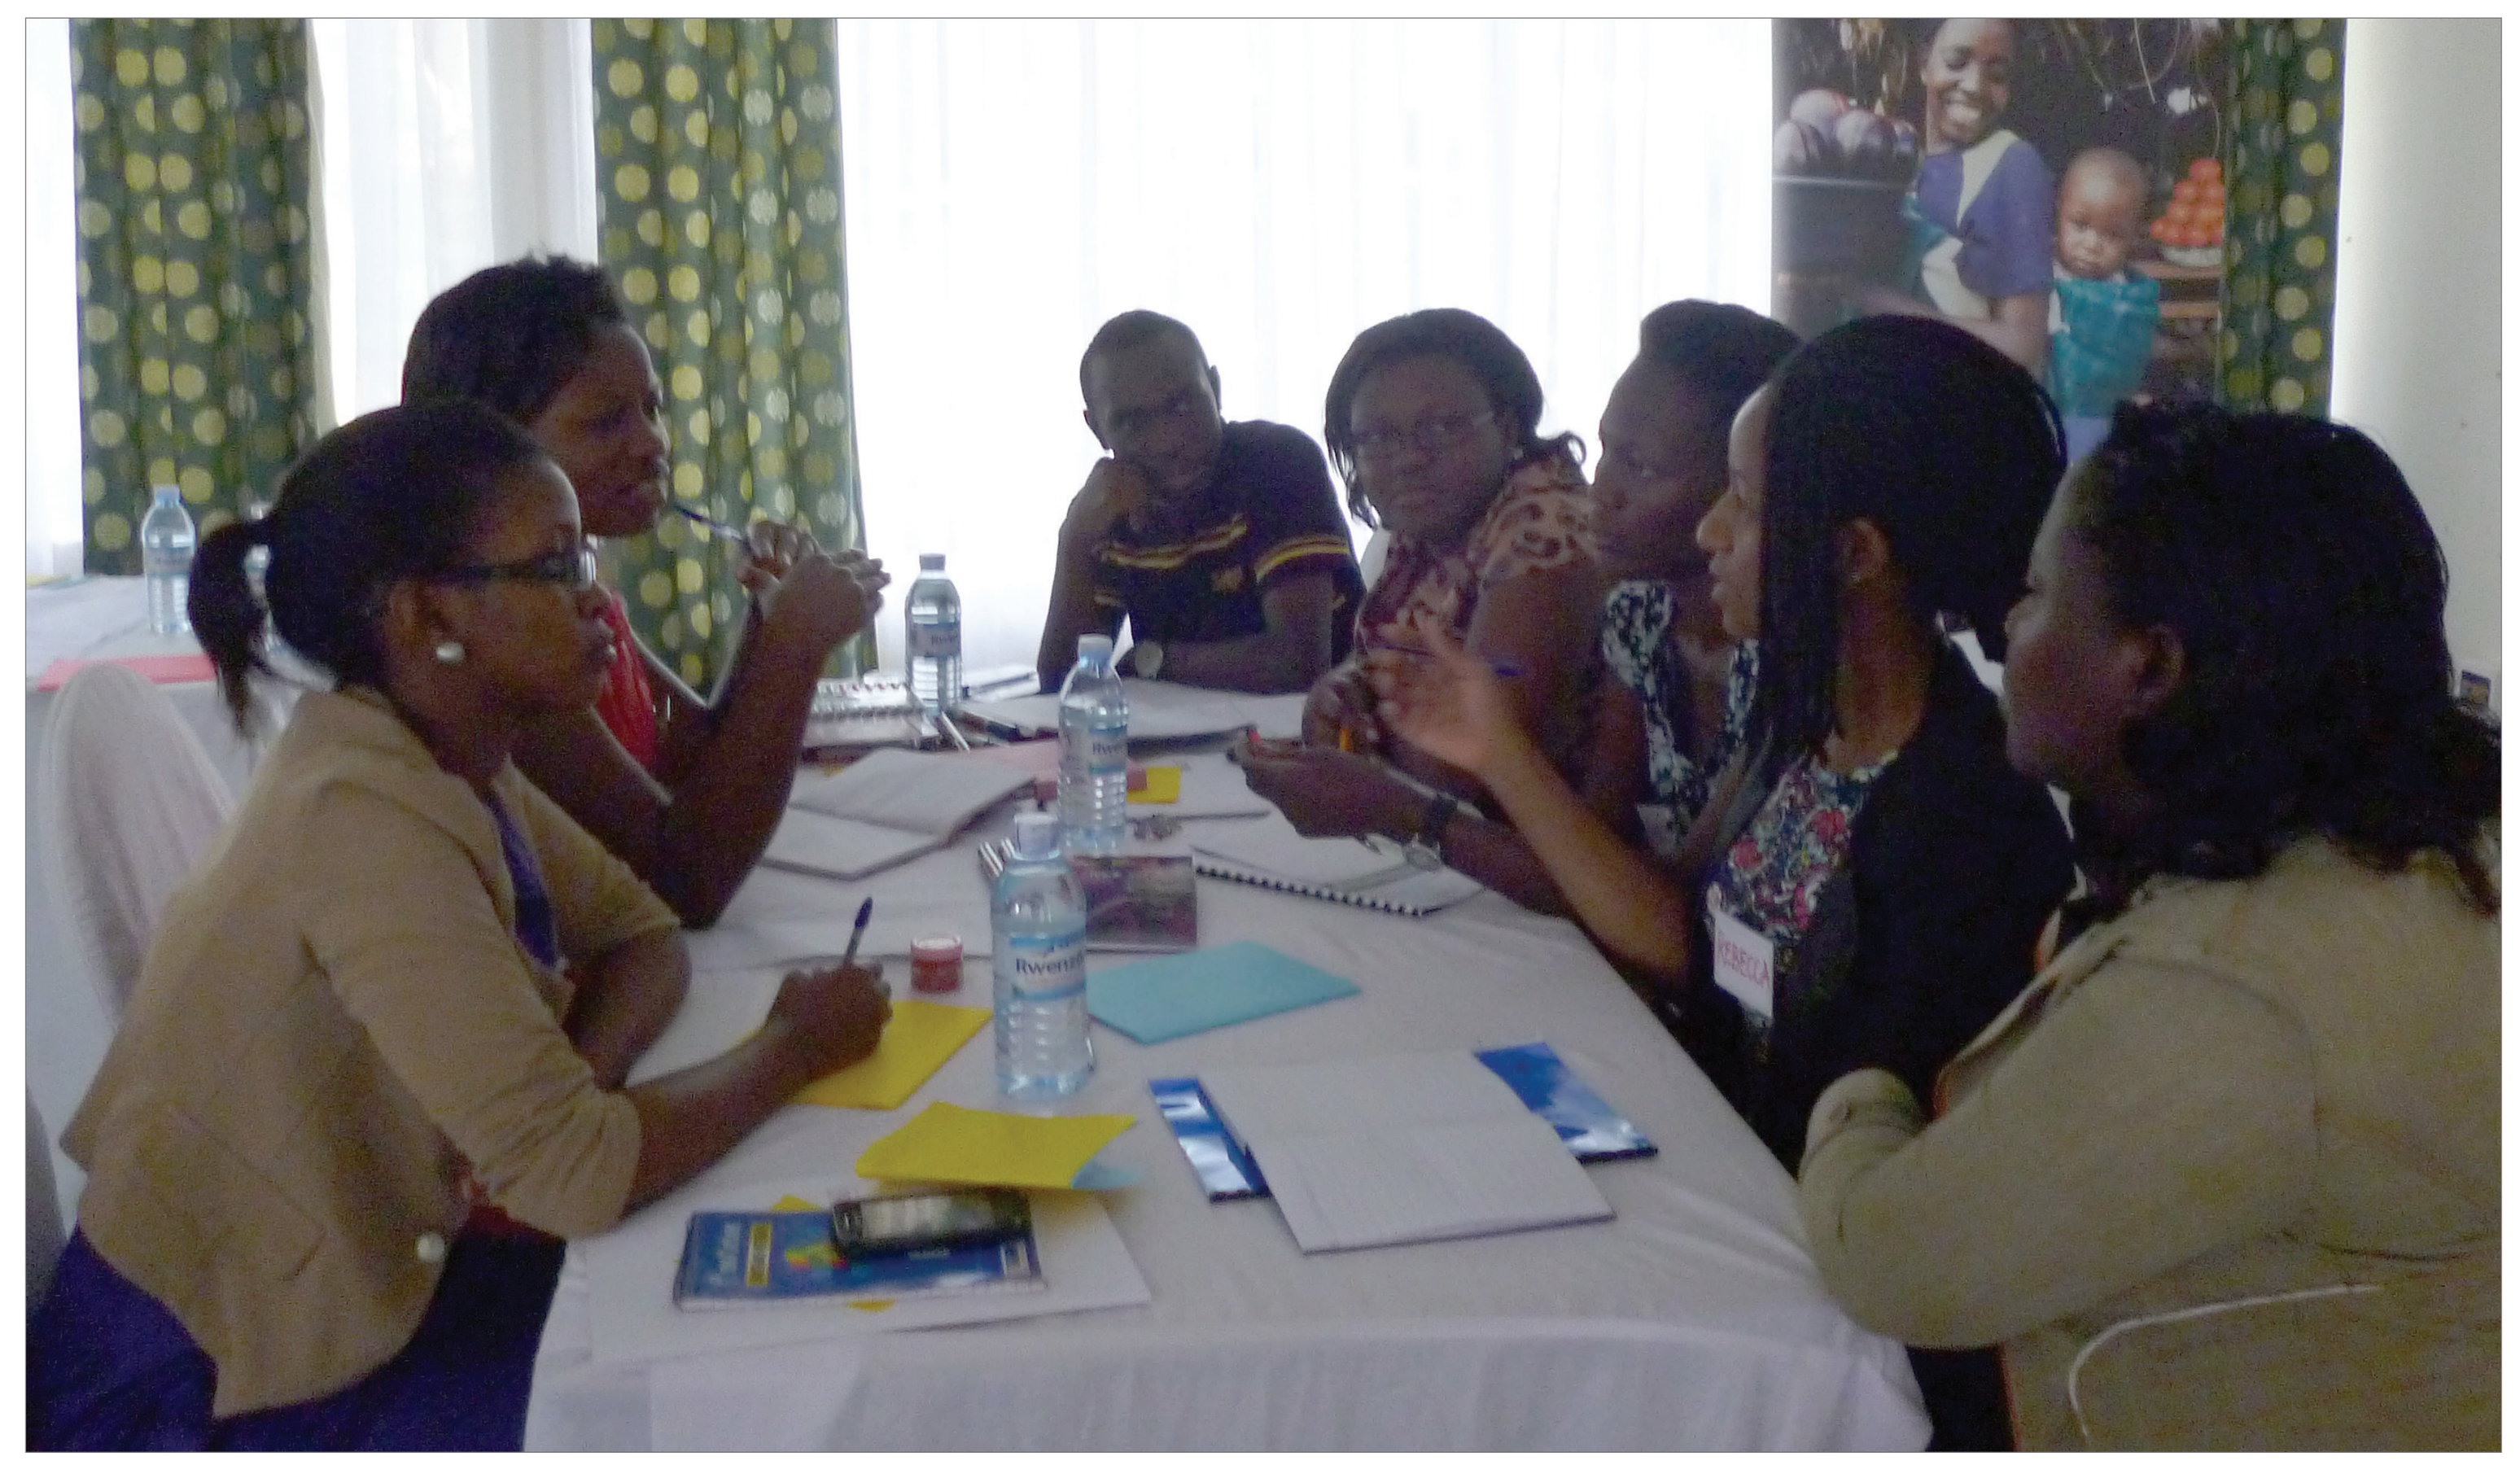

Roundtable discussions were held with fellows and mentors at retreats.

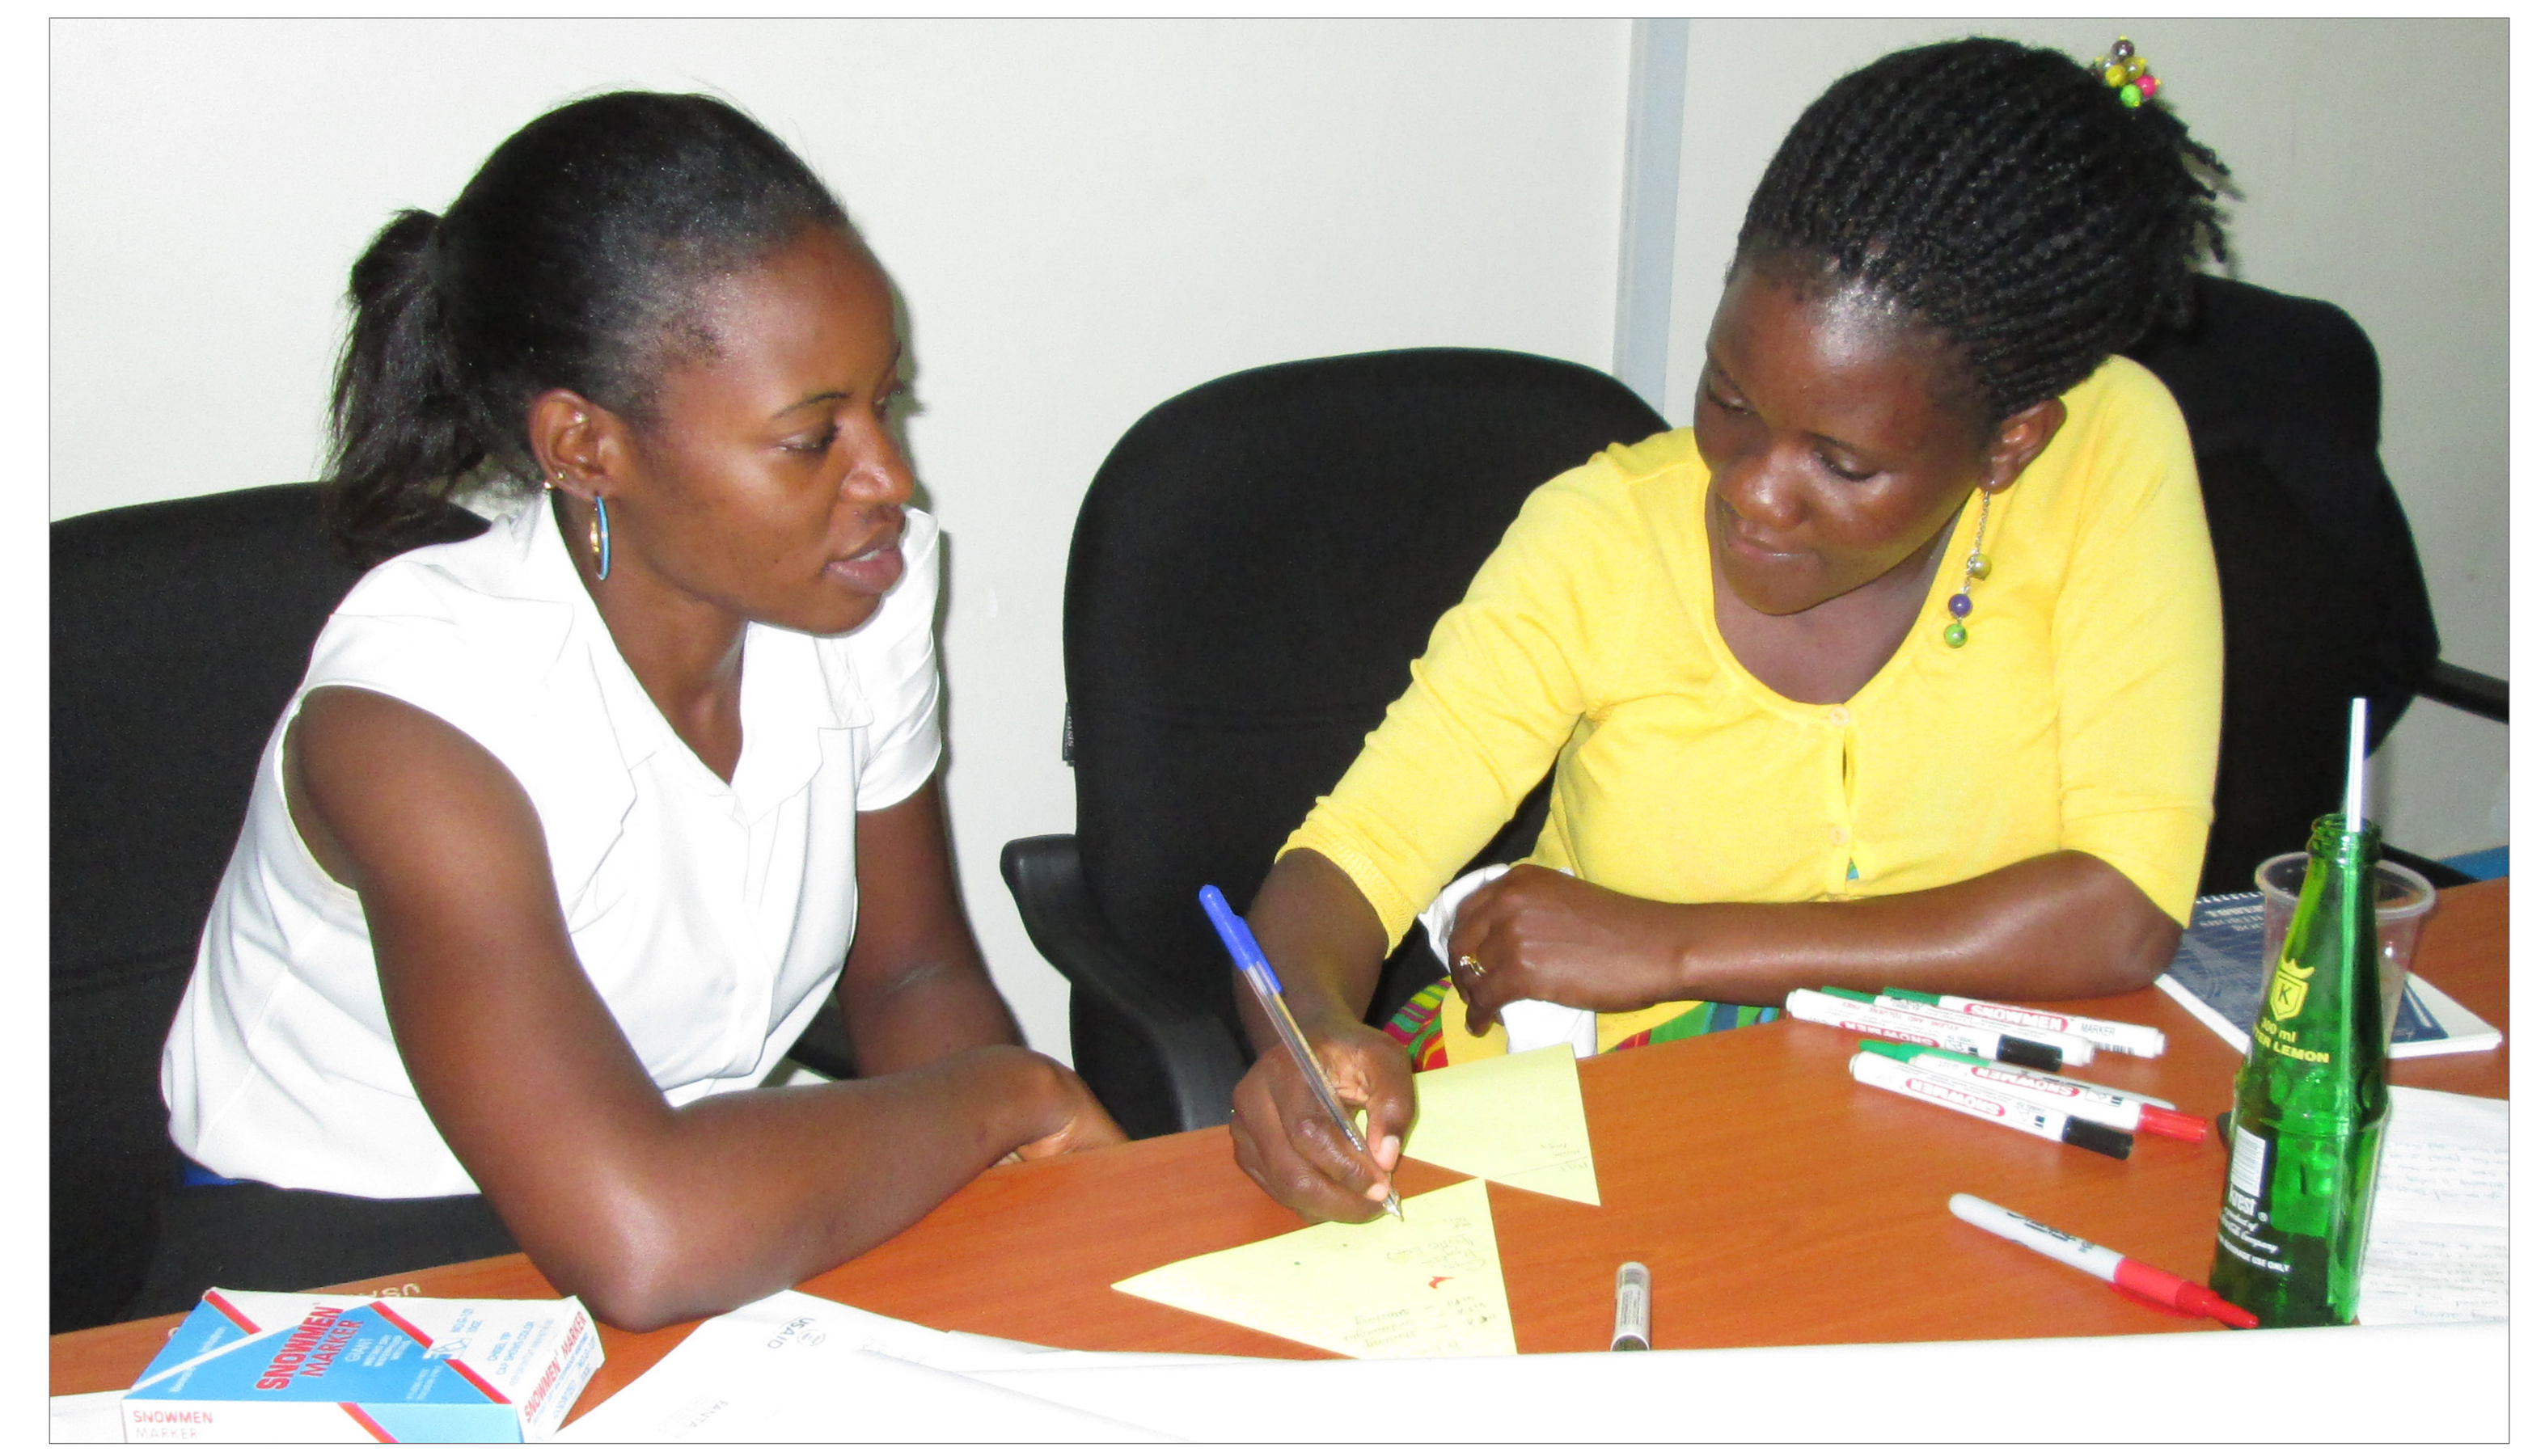

Fellows working on their UNF projects during a retreat.

FINDINGS AND WAY FORWARD

The UNF was a well-received and successful program; the eight fellows all found full-time employment or enrolled in advanced degree programs in nutrition within four months of program completion. While the UNF program provided funding and management, other stakeholders also played a key role. Host organizations provided mentorship and supervision as well as the practical experience necessary for young professionals to advance their careers, while other experts shared knowledge and expertise with fellows at professional development workshops and retreats throughout the program.

Feedback from fellows and host organizations guided improvements for the second year of the program, including longer-term placements and additional professional development opportunities. The 2015–2016 UNF program has grown—12 fellows were selected and placed at host organizations for 18 months. Additional professional development workshops and retreats will be held and relevant e-learning courses were added to the UNF program requirements. In order to better monitor and evaluate the program, fellows completed a skills and competencies self-evaluation to rank their core nutrition competencies and professional skills at the beginning of the program, and will do so again midway through and upon completion of the program. Results from this evaluation will be used to inform the content of workshops and retreats throughout the program. FANTA staff will visit fellows at their host organizations to ensure they are in positive learning environments and learn how to best support the fellows and the host organizations.

Learn more about the UNF program at www.fantaproject.org.

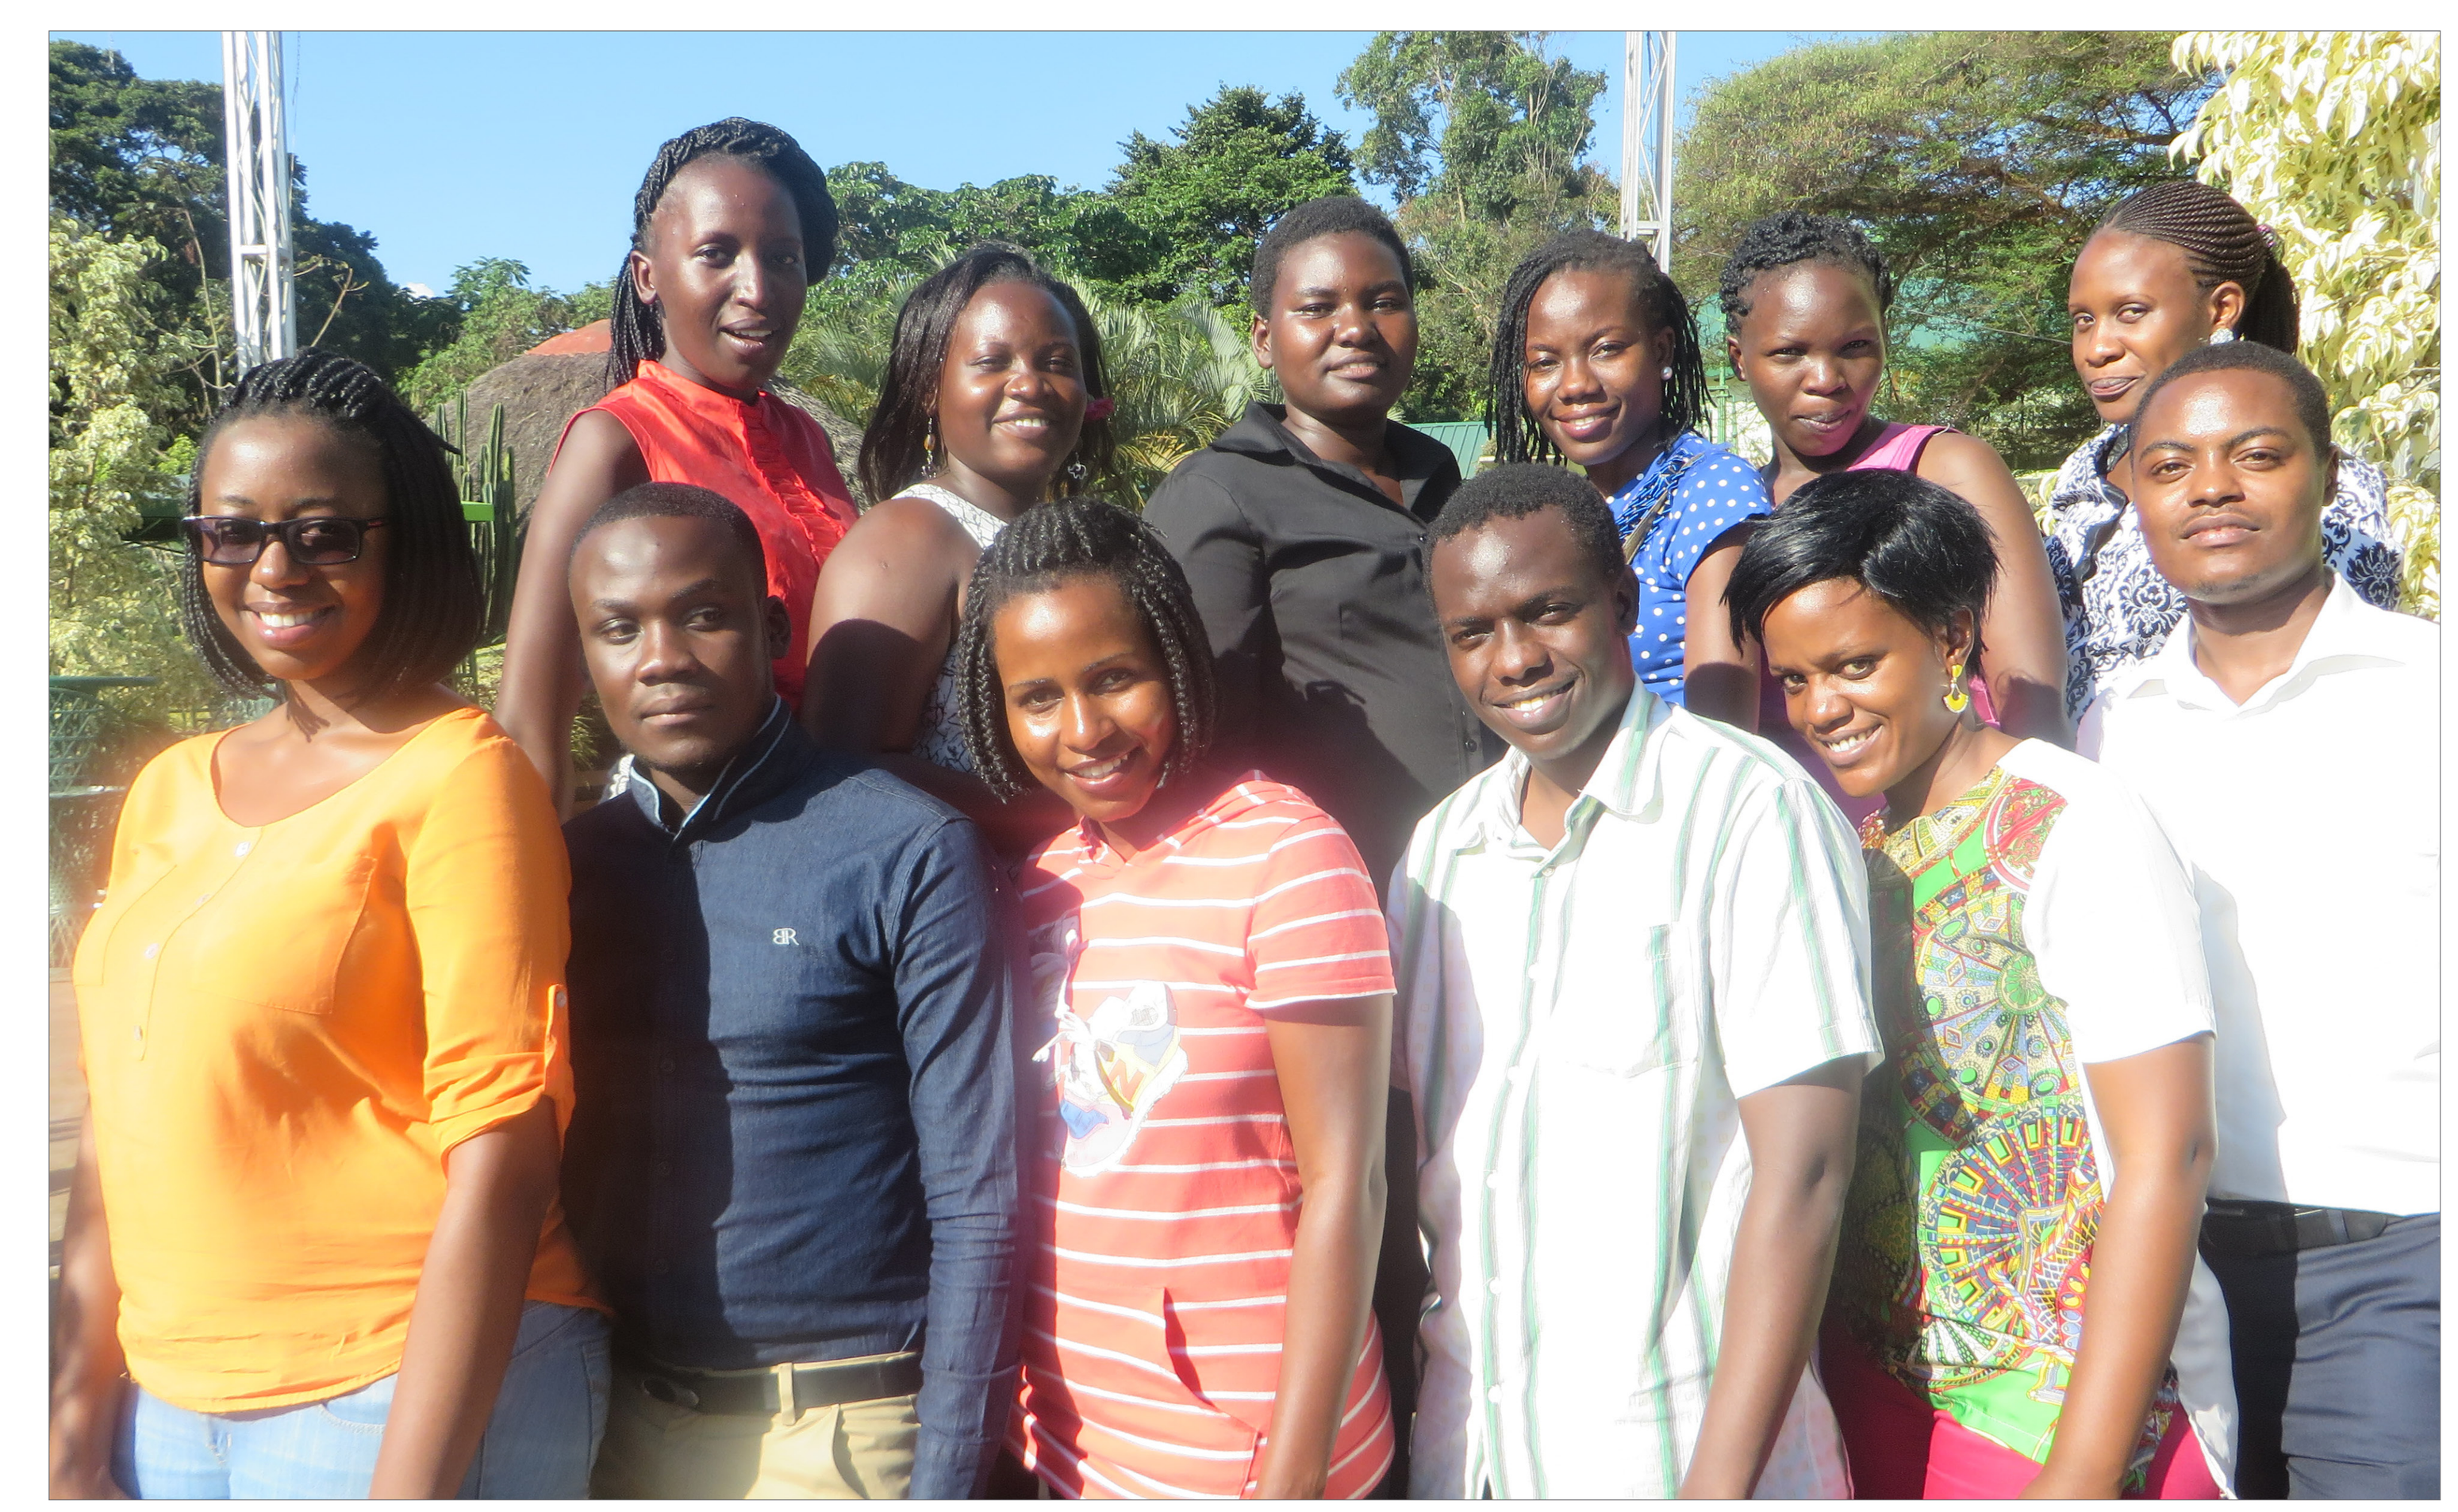

UNF fellows 2014–2015